Deep Learning-Based Human Activity Recognition for Continuous Activity and Gesture Monitoring for Schizophrenia Patients with Negative Symptoms

Daniel Umbricht[1*], Wei-Yi Cheng[2], Florian Lipsmeier[1], Atieh Bamdadian[1], Paul Tamburri[2], Michael Lindemann[1,3*]

[1] Roche Innovation Center Basel, F. Hoffmann-La Roche Ltd. Basel, Switzerland [2] Roche Innovation Center New York, Roche TCRC Inc, New York, USA [3] Baden-Wuerttemberg Cooperative State University, Loerrach, Germany;
* Corresponding authors

1. Introduction

- **Diminished expression and amotivation/avolition are key factors of the negative symptoms of schizophrenia. Among them, amotivation represents the critical component driving functional outcome [1].**
- **Quantified evaluation and continuous monitoring of behavior and symptoms associated with negative symptoms has been a challenge in clinical trials.**
- **Recent development in wearable sensor technology provides new opportunities to tackle these problems.**
- **However, to connect the sensor data with clinical observations and measures of motivated behavior still requires development of advanced analytics and real-world validations.**

2. Trial design and data collection

- Thirty-three schizophrenia patients with moderate negative symptoms were recruited in a 3-way cross-over proof of mechanism study (Roche study BP29904; Table 1).
- Of these, 31 patients were provided with a GeneActiv© wrist-worn actigraphy device to record actigraphy data for 15 weeks.
- Motivated behavior was assessed with an effort choice task (Fig 1a) [2]. % of high effort choices was the outcome measure.
- Negative symptoms were rated with the Brief Negative Symptom Scale (BNSS) and Positive and Negative Syndrome Scale (PANSS).
- The actigraphy device recorded acceleration of wrist movement at 20Hz. Patient adherence was acceptable. Median collected monitoring data per patient was 1,859 hours (about 11 weeks or 74%, Fig 1b).
- For all analyses, actigraphy data obtained over the last 5 days of the placebo period and assessments at the end of the placebo period were used.

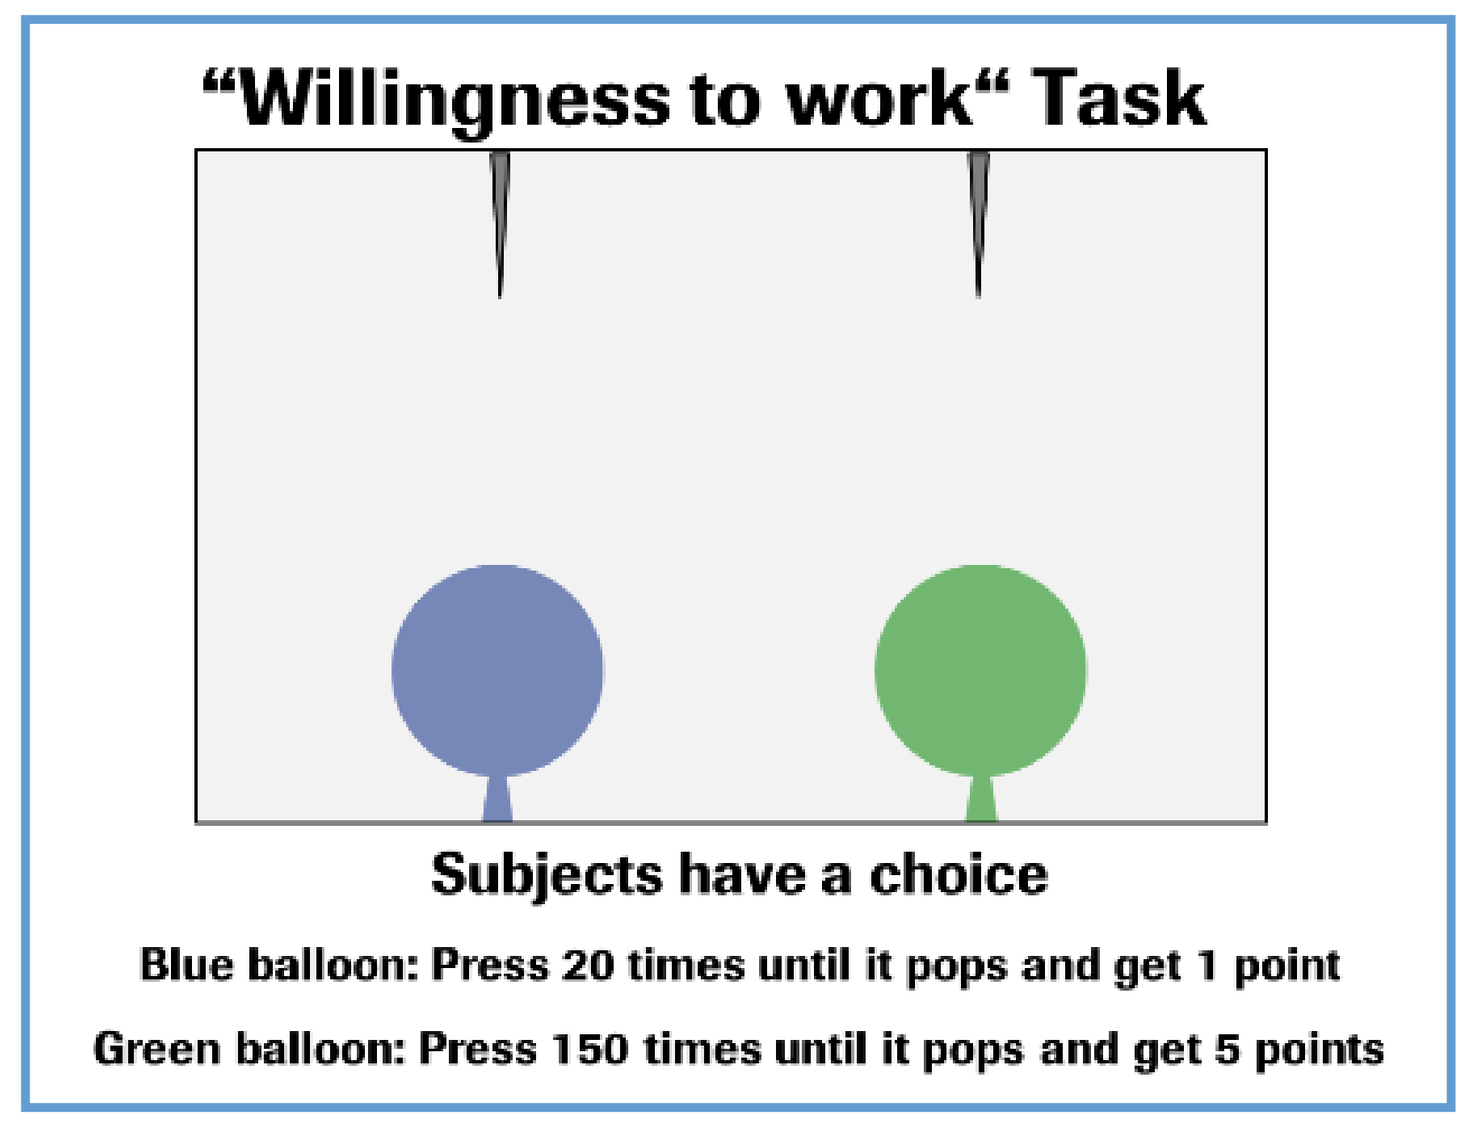
“Willingness to work“ Task
Subjects have a choice
Blue balloon: Press 20 times until it pops and get 1 point
Green balloon: Press 150 times until it pops and get 5 points

Fig 1a. Effort choice task

Parameter	Cohort statistics
N	33
Age (y)	36.6 ± 7.0
Race	21 Black, 9 White, 3 Asian
Gender (% male)	91
BNSS total score	36.0 ± 3.6
PANSS NSFS	22.8 ± 1.4
PANSS PSFS	19.4 ± 1.8

Table 1. Patient characteristics at baseline

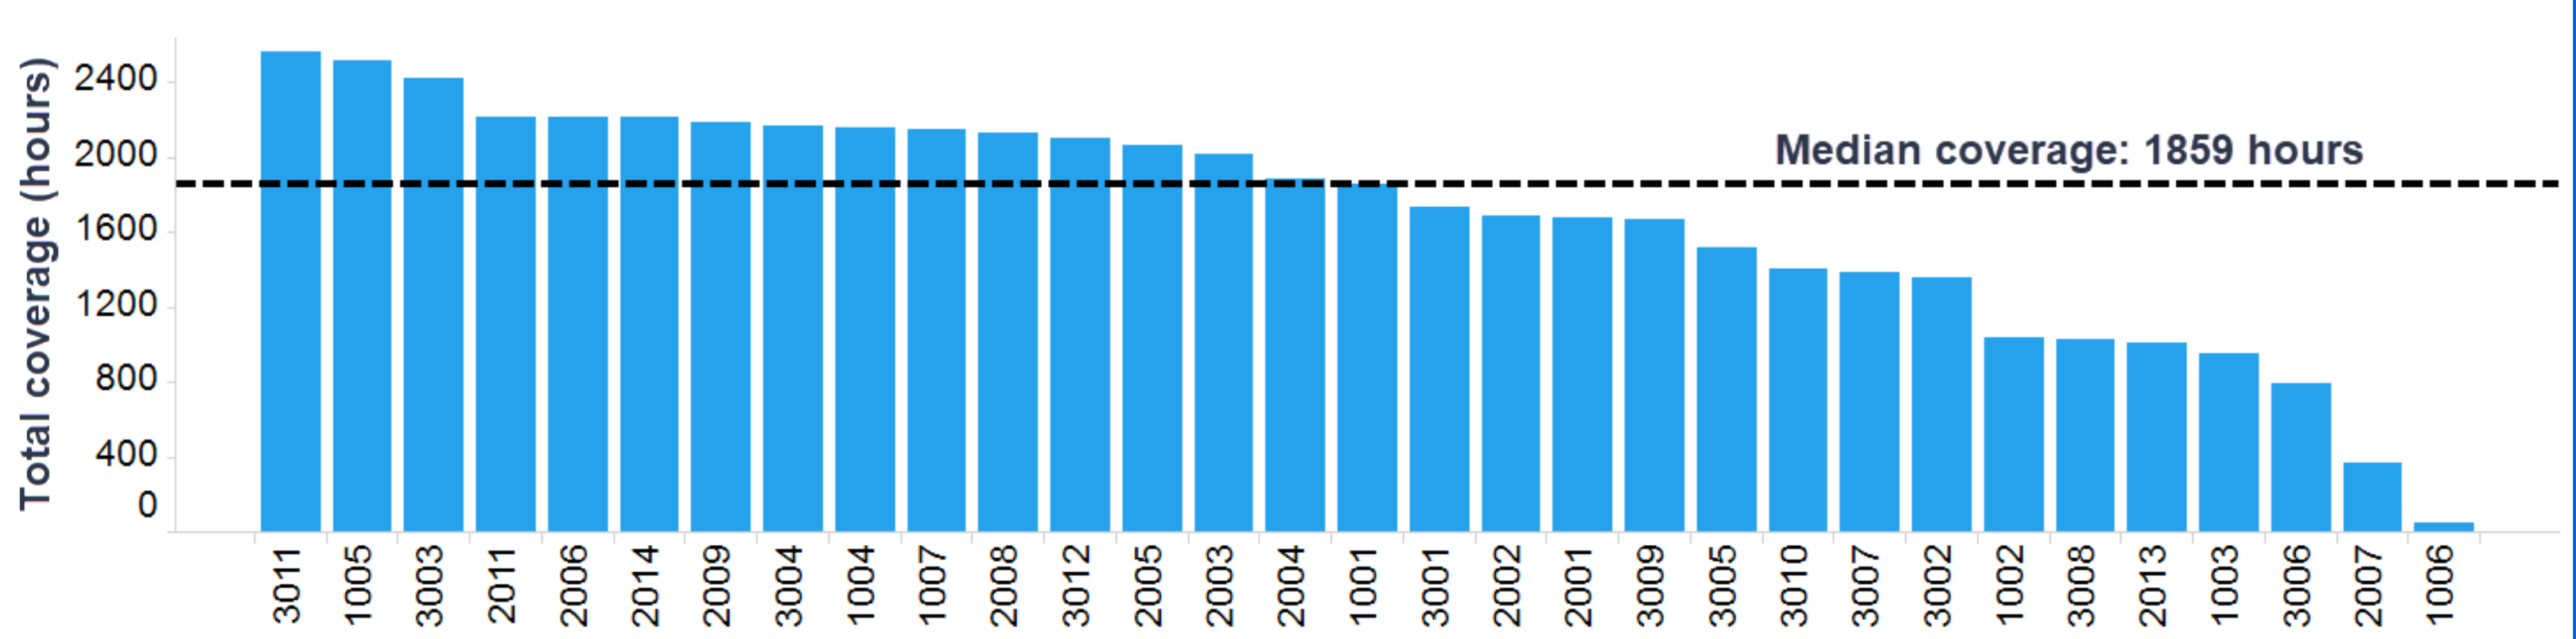
Total coverage (hours)
2400
2000
1600
1200
800
400
0
Median coverage: 1859 hours
3011 1005 3003 2011 2006 2014 2009 3004 1004 1007 2008 3012 2005 2003 2004 1001 3001 2002 2001 3009 3005 3010 3007 3002 1002 3008 2013 1003 3006 2007 1006

Fig 1b. Actigraphy data per patient

3. Human Activity Recognition (HAR) and Gesture Feature Derivation

- We trained a 9-layer convolutional recurrent neural network (Fig 2a) using two public annotated data sets containing wrist-worn acceleration data to infer the subjects' activities [3][4].
- The trained human activity recognition (HAR) model was tested with held-out validation data and internally collected sensor data from previous studies [5]. More than 95% of accuracy to separate the ambulatory (walking, stairs, cycling, jogging) from stationary activities (sitting, standing, lying down, hand work) was achieved (Fig 2b).
- From the predicted activities, we derived for each patient the activity ratio [= the total active time divided by the monitoring time].
- We further inferred the gesture events and gesture features based on the activities and acceleration signal using an empirically defined threshold on accelerometer signal from the wrist [6].

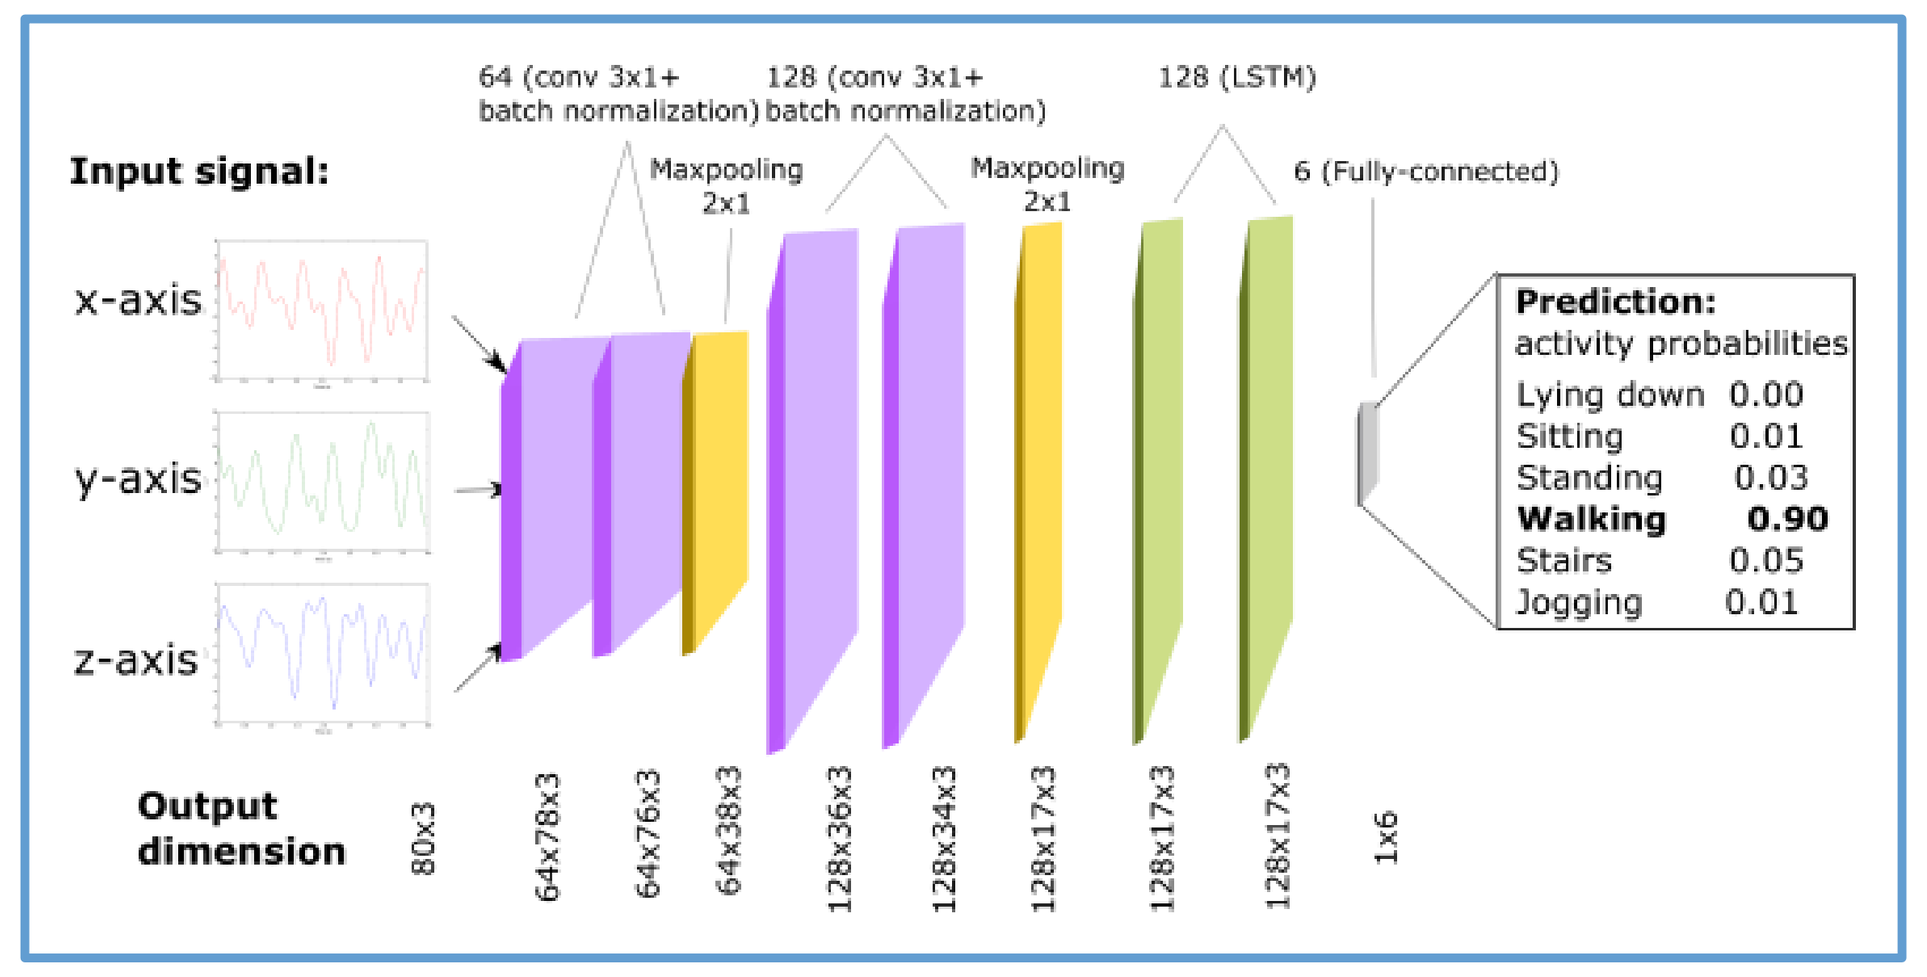
Input signal:
x-axis
y-axis
z-axis
64 (conv 3x1+ batch normalization)
128 (conv 3x1+ batch normalization)
Maxpooling 2x1
Maxpooling 2x1
128 (LSTM)
6 (Fully-connected)
Prediction:
activity probabilities
Lying down 0.00
Sitting 0.01
Standing 0.03
Walking 0.90
Stairs 0.05
Jogging 0.01
Output dimension
80x3
64x78x3
64x76x3
64x38x3
128x36x3
128x34x3
128x17x3
128x17x3
128x17x3
1x6

Fig 2a. 9-layer convolutional neural network

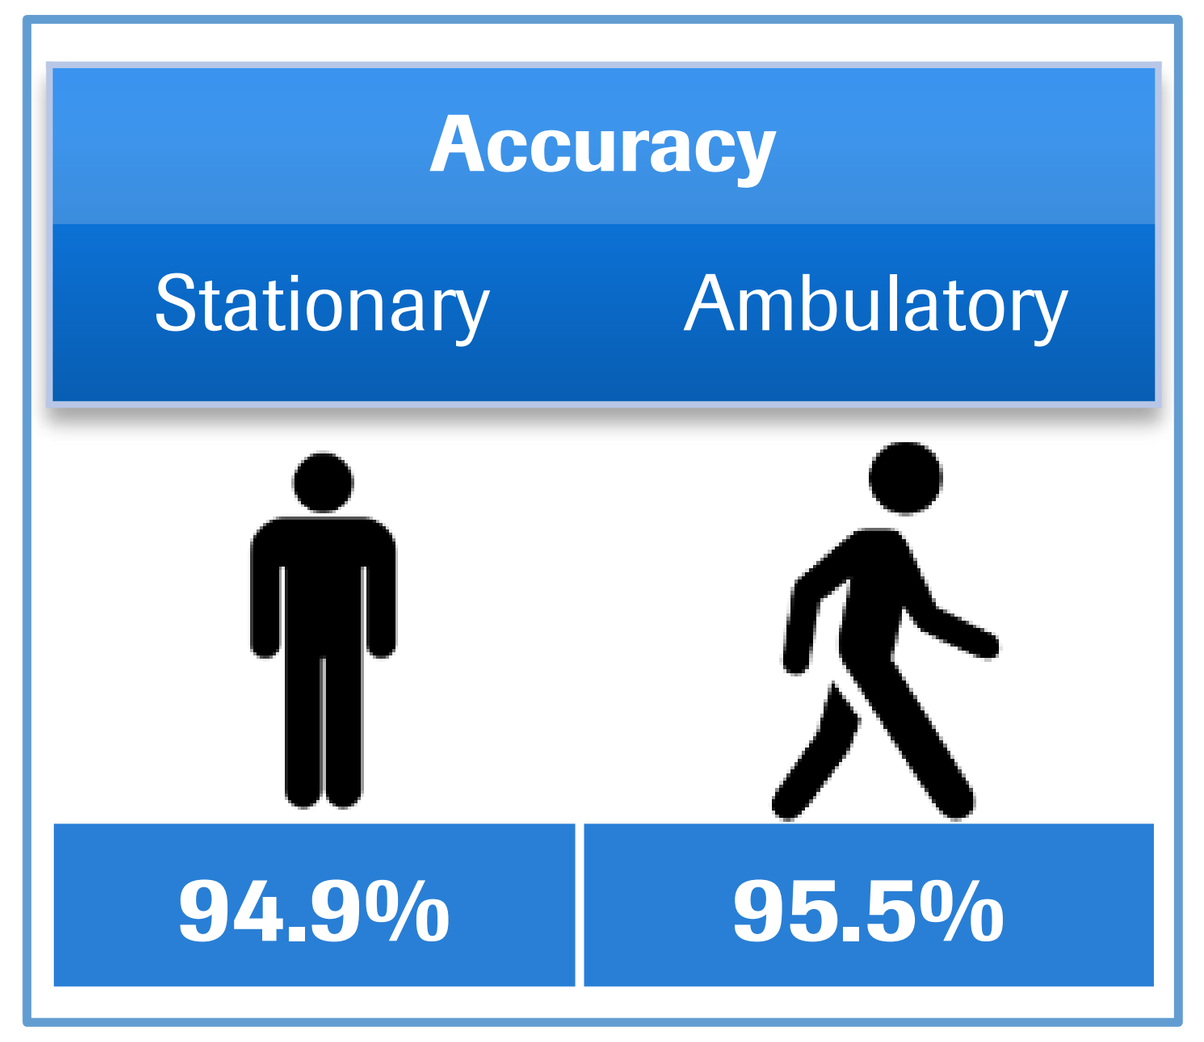
Accuracy
Stationary
Ambulatory
94.9%
95.5%

Fig 2b. Prediction accuracy on labeled trial data from previous study

4. Activity ratio correlates with high effort choice

- The activity time ratio significantly correlated with the % high effort choice at the end of the placebo period (Spearman r = 0.58, P value = 0.002, Fig 3), indicating an association of avolition and lower activity in daily life.

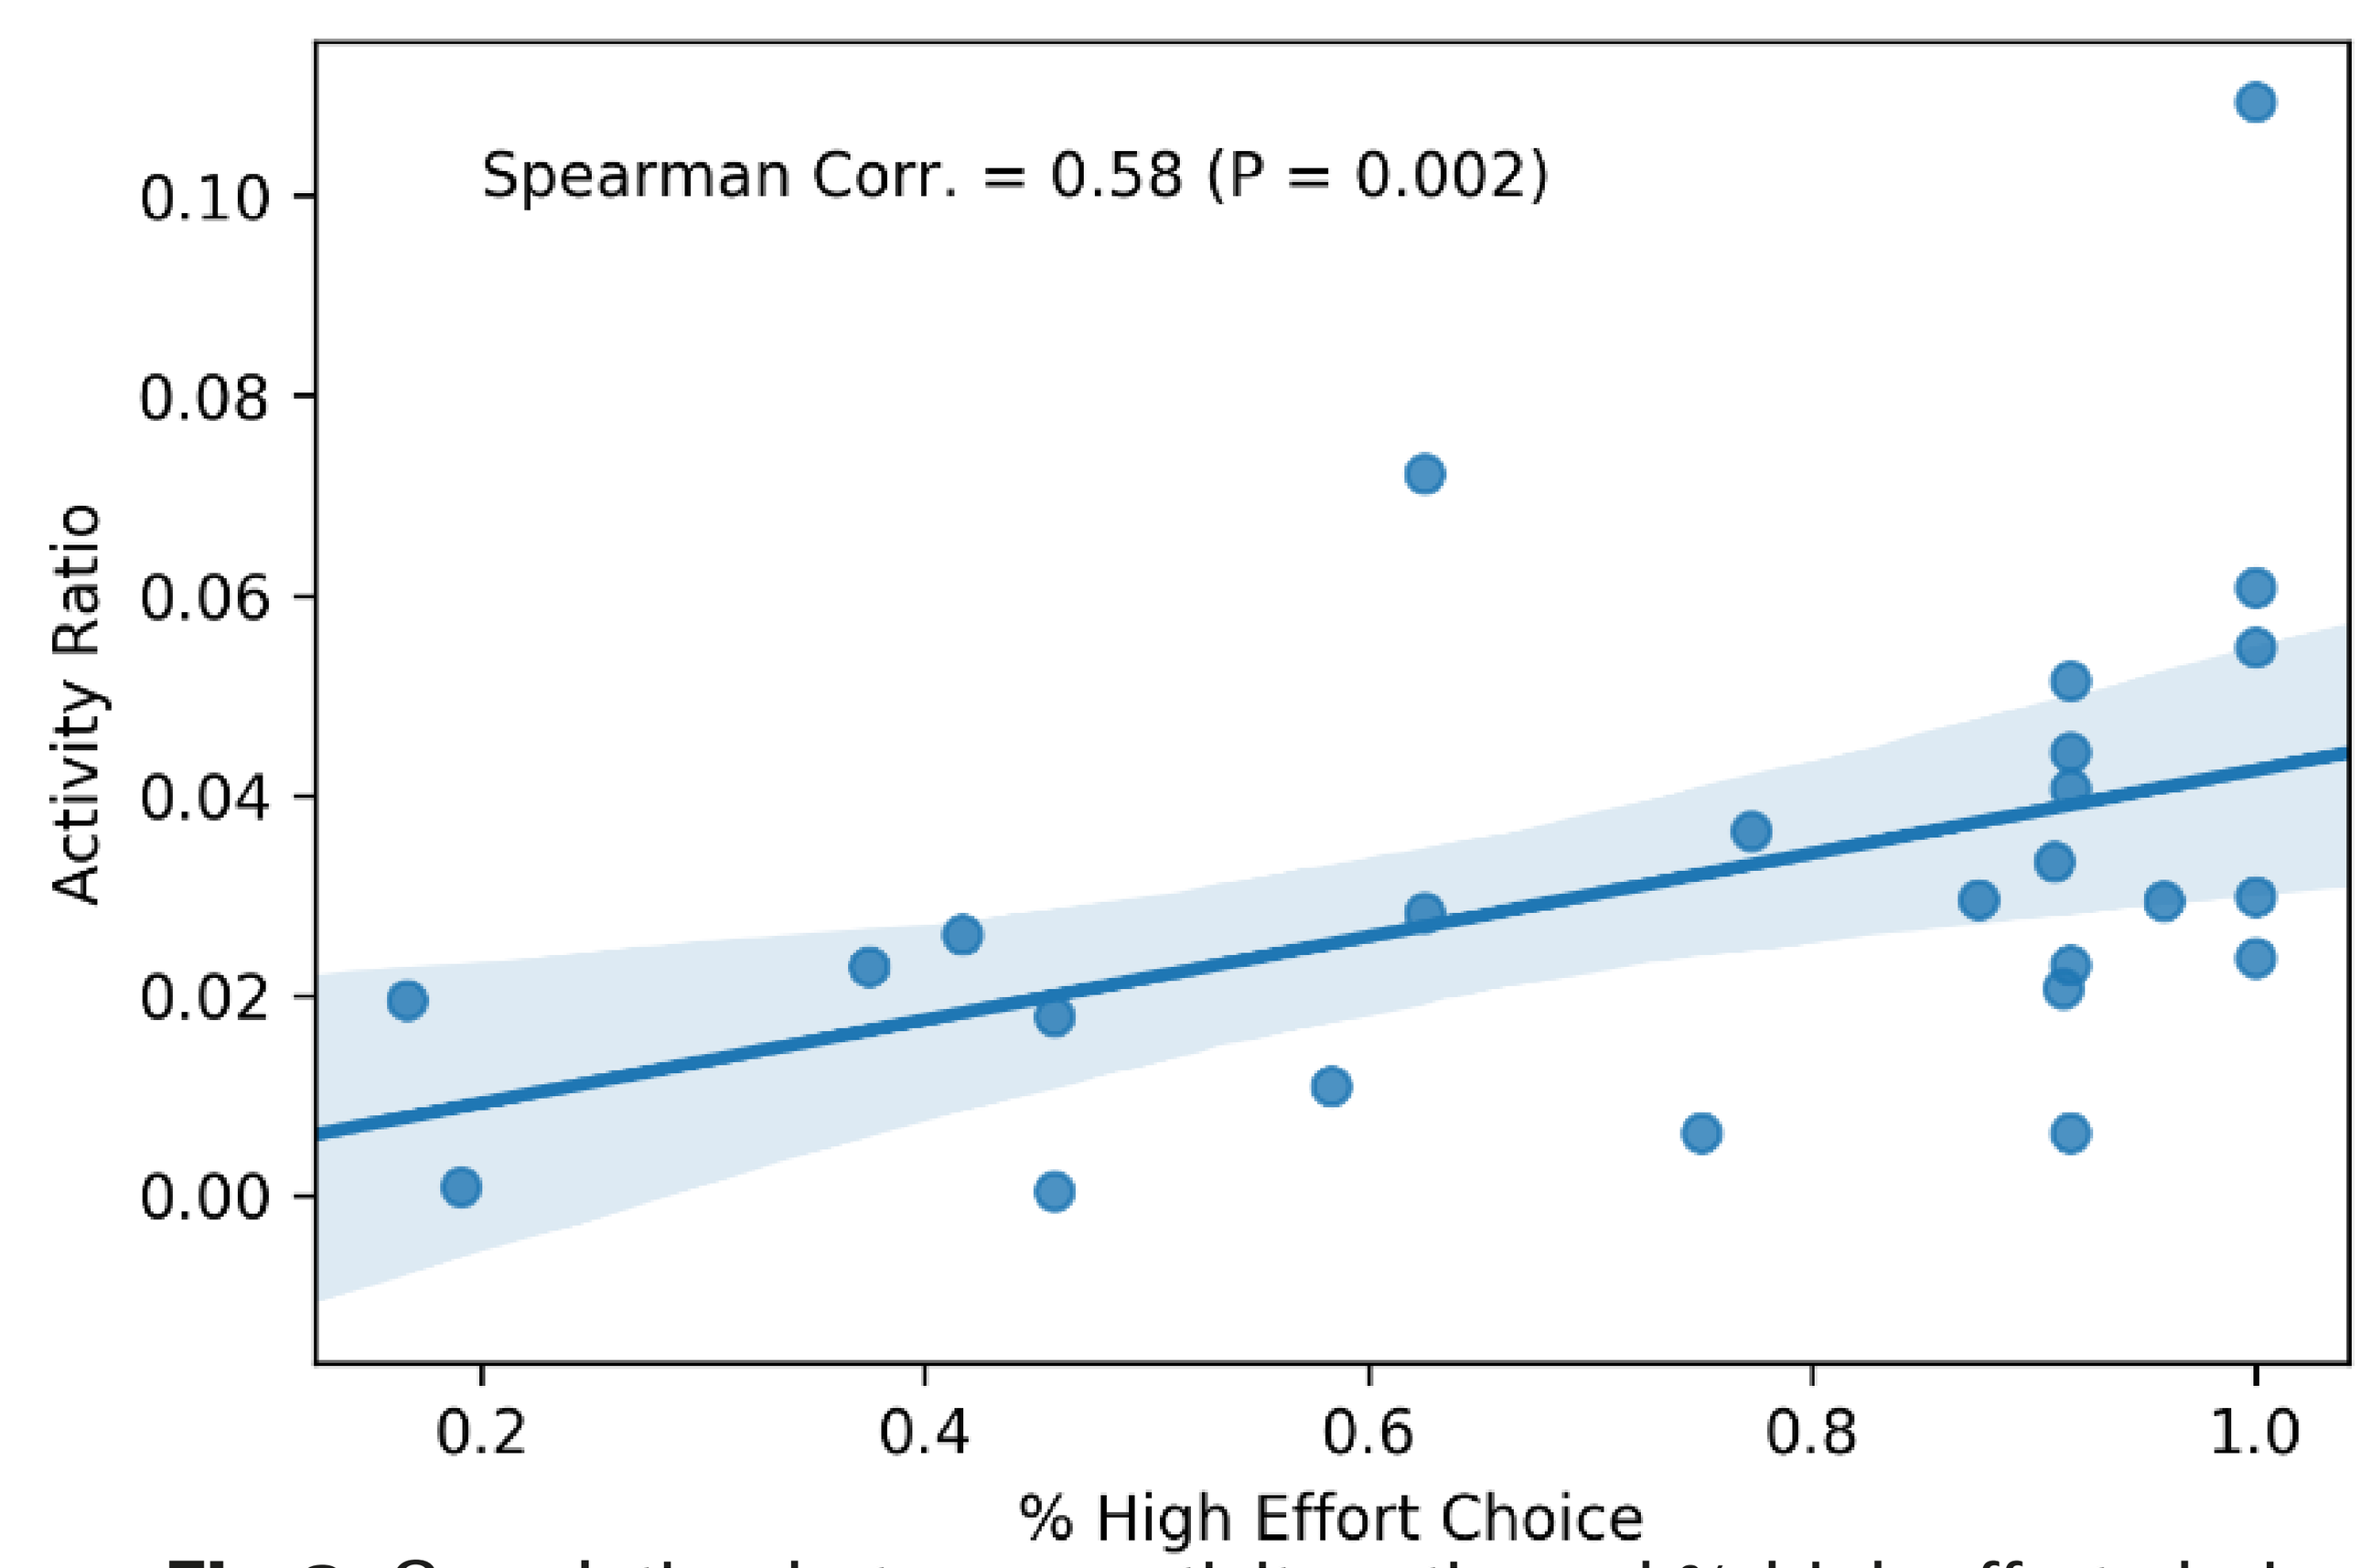
Spearman Corr. = 0.58 (P = 0.002)
Activity Ratio
0.10
0.08
0.06
0.04
0.02
0.00
0.2
0.4
0.6
0.8
1.0
% High Effort Choice

Fig 3. Correlation between activity ratio and % high effort choice

5. Gesture features correlate with BNSS

- Median daily gesture counts correlated negatively with BNSS total score (Spearman r = -0.44, P value = 0.03, Fig 4a), specifically with the diminished expression sub-score (Spearman r = -0.42, P value = 0.03, Fig 4b).
- Gesture features correlated with BNSS total score and diminished expression subscores (Table 2).

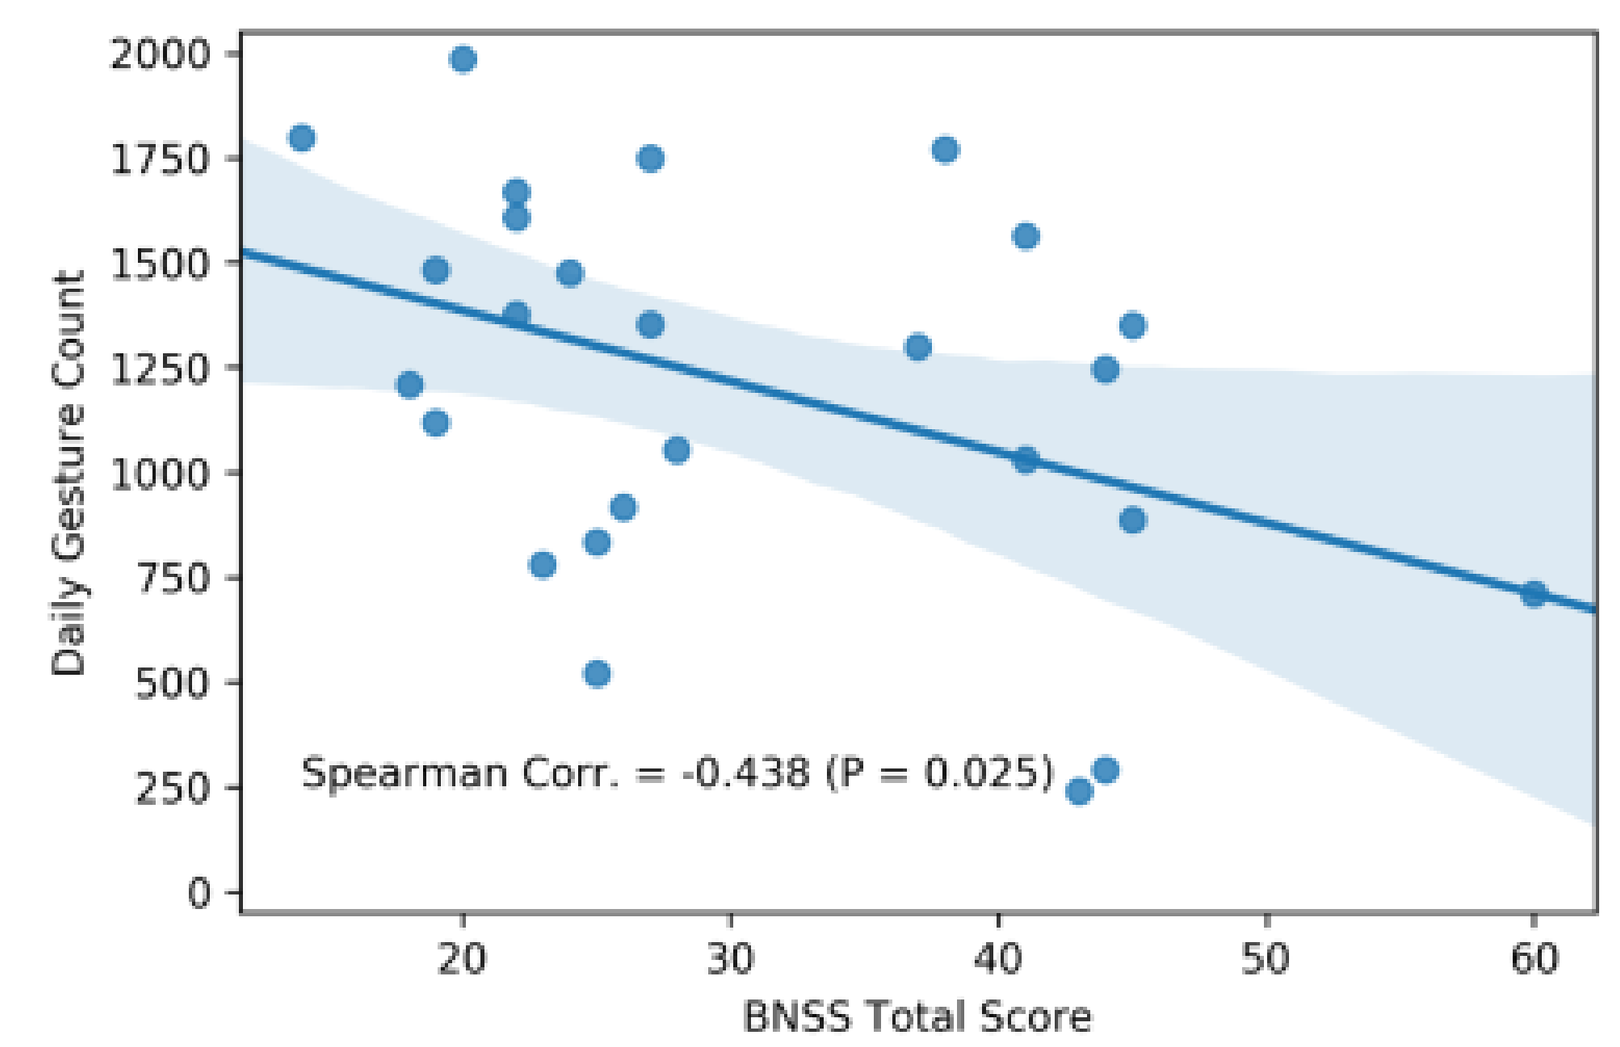
Daily Gesture Count
Spearman Corr. = -0.438 (P = 0.025)
BNSS Total Score

Fig 4a. Daily gesture count and BNSS total score

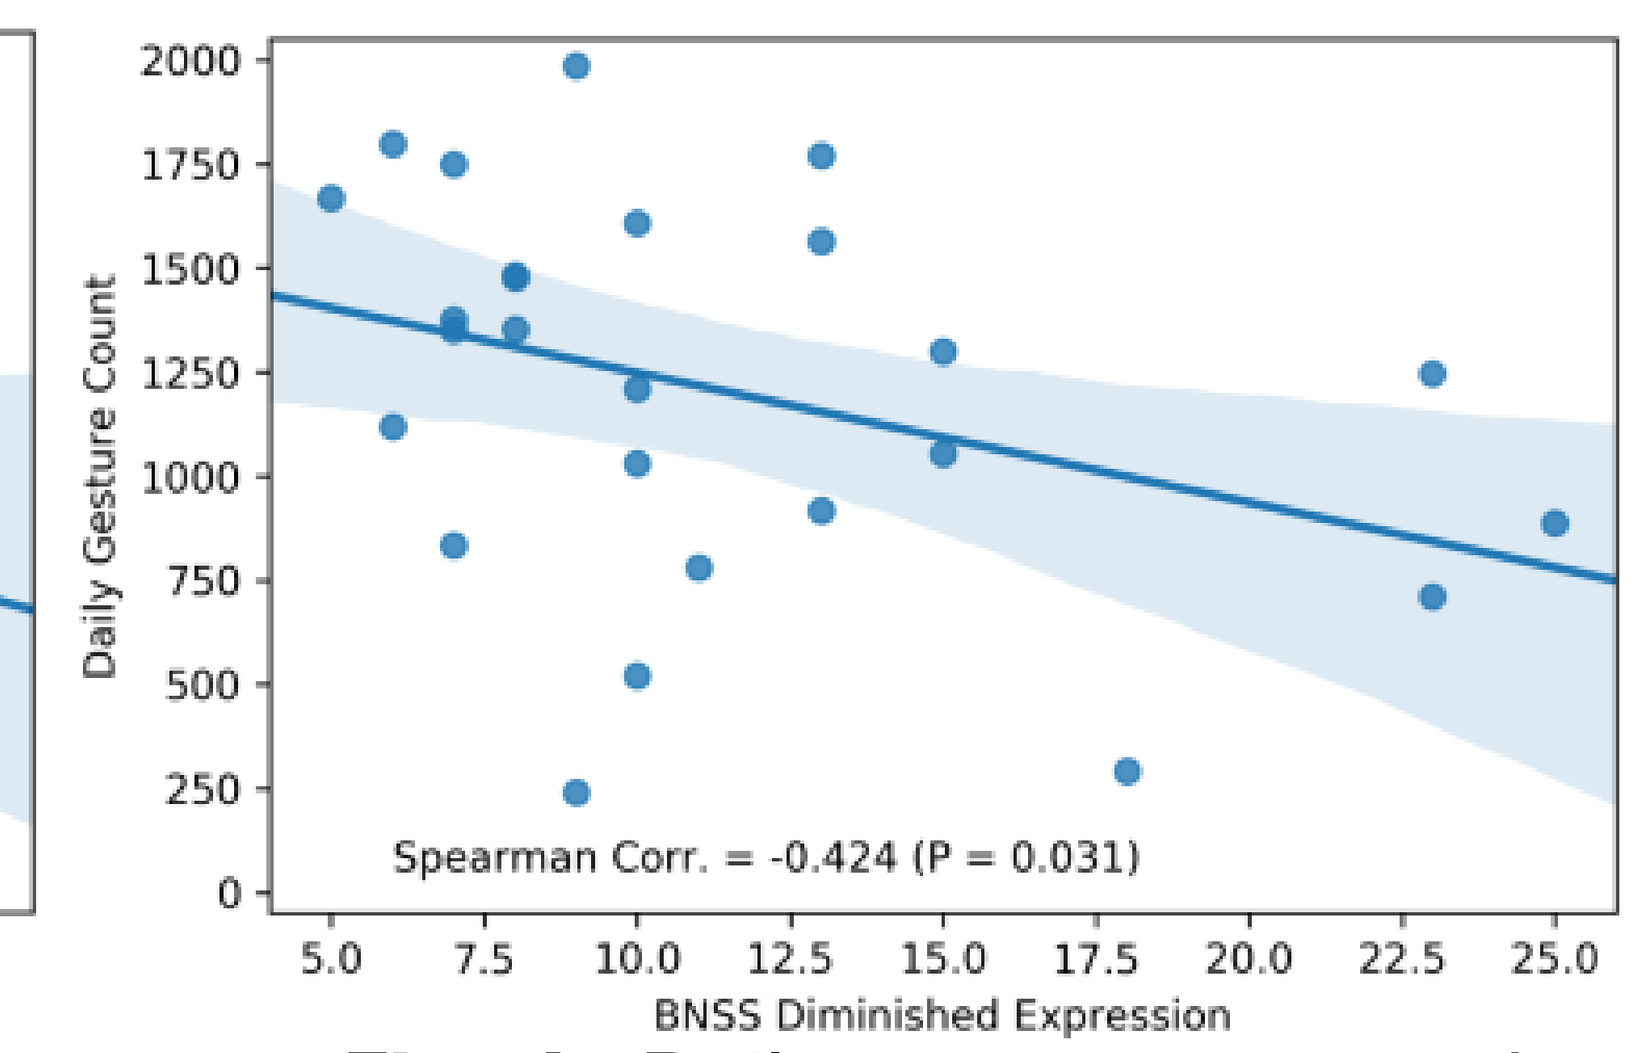
Daily Gesture Count
Spearman Corr. = -0.424 (P = 0.031)
BNSS Diminished Expression

Fig 4b. Daily gesture count and BNSS diminished expression

	Activity ratio	Gesture power	Gesture count
BNSS Apathy Index	-0.025	-0.178	-0.277
BNSS Diminished Expression	-0.210	**-0.423***	**-0.424***
BNSS Total Score	-0.080	-0.312	**-0.438***
PANSS NSFS	-0.256	0.080	-0.251
PANSS PSFS	-0.149	0.164	-0.026

Table 2. Correlations between activity and gesture features with clinical scores; * $p<0.05$

6. Discussion and Outlook

- **The results demonstrate the feasibility of using wrist-worn actigraphy for continuously monitoring clinically-relevant behavior in a clinical trial setting.**
- **Activity and gesture features derived from a HAR model were associated with motivated behavior and expressive deficits, respectively, supporting the validity of activity and gesture-based digital biomarkers for negative symptoms.**
- **The activity and gesture features derived from HAR model shows promise to be used as biomarker for drug development**.

References

[1] G. Foussias *et al.*, “Negative Symptoms in Schizophrenia: Avolition and Occam's Razor,” Schizophrenia Bulletin, Vol. 36, Issue 2, 2010, pp359-369.
[2] J.M. Gold *et al.*, “Negative Symptoms of Schizophrenia Are Associated with Abnormal Effort-Cost Computations,” Biol Psychiatry 2013; 74:130-136.
[3] A. Reiss *et al.*, “Introducing a New Benchmarked Dataset for Activity Monitoring.” The 16th IEEE International Symposium on Wearable Computers, 2012.
[4] D. Anguita *et al.*, “A Public Domain Dataset for Human Activity Recognition Using Smartphones.” 21th European Symposium on Artificial Neural Networks, Computational Intelligence and Machine Learning, ESANN 2013. Bruges, Belgium 24-26 April 2013.
[5] X. Montalban *et al.*, “FLOODLIGHT: Remote Self-Monitoring Is Accepted by Patients and *Provides* Meaningful, Continuous Sensor-Based Outcomes Consistent With and Augmenting Conventional In-Clinic Measures,” (P4.382)
[6] A. Rai *et al.*, “Zee: Zero-Effort Crowdsourcing for Indoor Localization,” MobiCom'12, August 22–26, 2012