Digital, high-frequency, long-term monitoring of motor and non-motor symptoms in Huntington's disease (HD)

Florian Lipsmeier (1), Wei-Yi Cheng (1), Detlef Wolf (1), Yan-Ping Zhang (1), Timothy Kilchenmann (1), Atieh Bamdadian (1), Anne Smith (2), Edward Wild (3), Scott Schobel (4), Christian Czech (1), Christian Gossens (1) and Michael Lindemann (1)

(1) Roche Pharmaceutical Research and Early Development, F. Hoffmann-La Roche Ltd., Basel, Switzerland (2) Ionis Pharmaceuticals Inc, Carlsbad, CA, USA (3) UCL Huntington's Disease Centre, Institute of Neurology, National Hospital for Neurology & Neurosurgery, Queen Square, London, UK (4) F. Hoffmann-La Roche Ltd., Basel, Switzerland

Summary

A study is underway to determine if smartphones and wrist-worn wearable technology can be used to measure HD symptoms in daily life using at-home tests and continuous movement tracking. The first results from the study show that most patients consistently completed the tests and that this may be a useful way of monitoring patients' symptoms, supplementing clinic visits.

Methods

Study Design

- Patients with HD in the ongoing ISIS 443139-CS2 clinical study (n=46) receive a smartphone (Galaxy S7; Samsung, Seoul, South Korea) and smartwatch (Moto G 360 2nd Gen Sport; Motorola, Chicago, USA) during screening.
- Patients first complete active tests using these tools in a teaching session in the clinic. The active tests are then done remotely at home and at follow-up clinical visits. For passive monitoring, patients are asked to carry the devices with them as they go about their daily activities (**Figure 1**).
- The sensor data is securely transferred via WiFi to Roche, where it is processed and analysed.
 - Adherence data examines the average number of active tests done per week and average hours/days per week of sensor data collected.
 - Sensor data will be compared to clinical data.
 - The relationship between UHDRS[1] item Maximal Chorea and respective sensor-derived features is assessed using Spearman's rank-order correlation.
 - Sensor data is compared to data collected from similar tests completed by independent HCs (N=35, age= 56.23 ± 7.83, 27 male/8 female).[2]
 - Wilcoxon tests were used for comparisons between HD patients and healthy controls.

Table 1: ISIS 443139-CS2 patient sub-population analyzed for this poster*

N	41
Age (years)	48.66 ± 9.95
Gender (male/female)	25/16
Independence scale	86.95 ± 8.13
TFC	11.10 ± 1.61
TMS	23.54 ± 12.56
Montreal Cognitive Assessment (total)	24.66 ± 3.24
SWR	74.39 ± 22.58
SDMT	34.24 ± 12.59

*Preliminary, non-cleaned data from patients with UHDRS and sensor data within the first 14 days of the study; Mean (± SD);
Data cut-off: Aug 6th, 2018.

Fig 1: Smartphone test activities and schedule

ACTIVE TESTS									
PROs		*Cognitive Tests*		*Upper Body Motor Tests*			*Stability and Gait*		
Daily Qs	EQ-5D-5L	SDMT	Word reading	Speeded Tapping	Draw a shape	Chorea	Balance	U-Turn	Walk
Daily	Weekly	Weekly	Weekly	Daily	Daily	Daily	Daily	Daily	Daily
Behavioral assessment	Behavioral assessment	SDMT	SWR	Finger Taps	Finger Taps Pronate/ Supinate-Hands	Maximal Chorea	Maximal chorea and Pull Test	Bradykinesia-body and Gait	Bradykinesia-body, Gait and tandem walking

PASSIVE MONITORING		
Activities of Daily Living		
Gait	Chorea	Activity Levels
Bradykinesia-body and Gait	Maximal Chorea	Bradykinesia-body and Gait

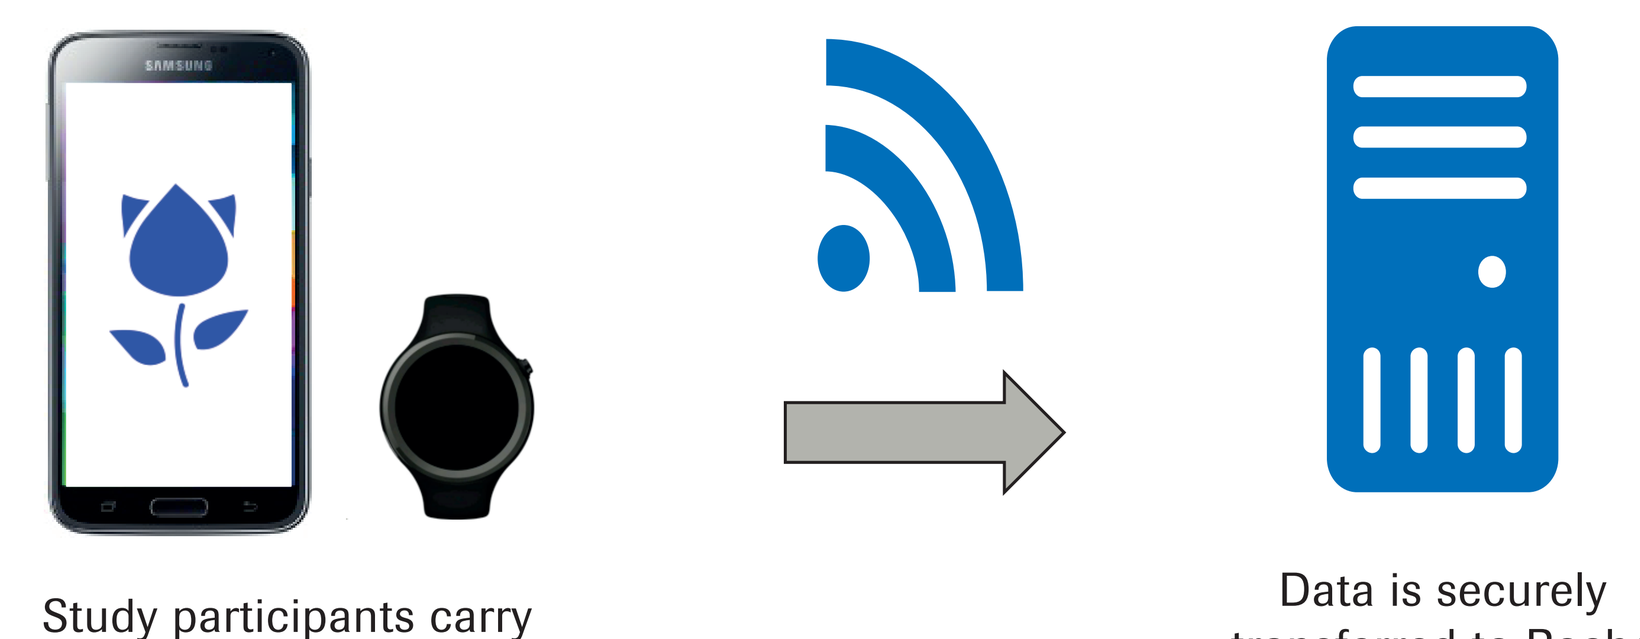
Study participants carry a smart phone and wear a smartwatch
Data is securely transferred to Roche via WiFi for analysis

Anchored to clinical assessments

Fig 2: Study participants display good adherence to remote digital monitoring assessments

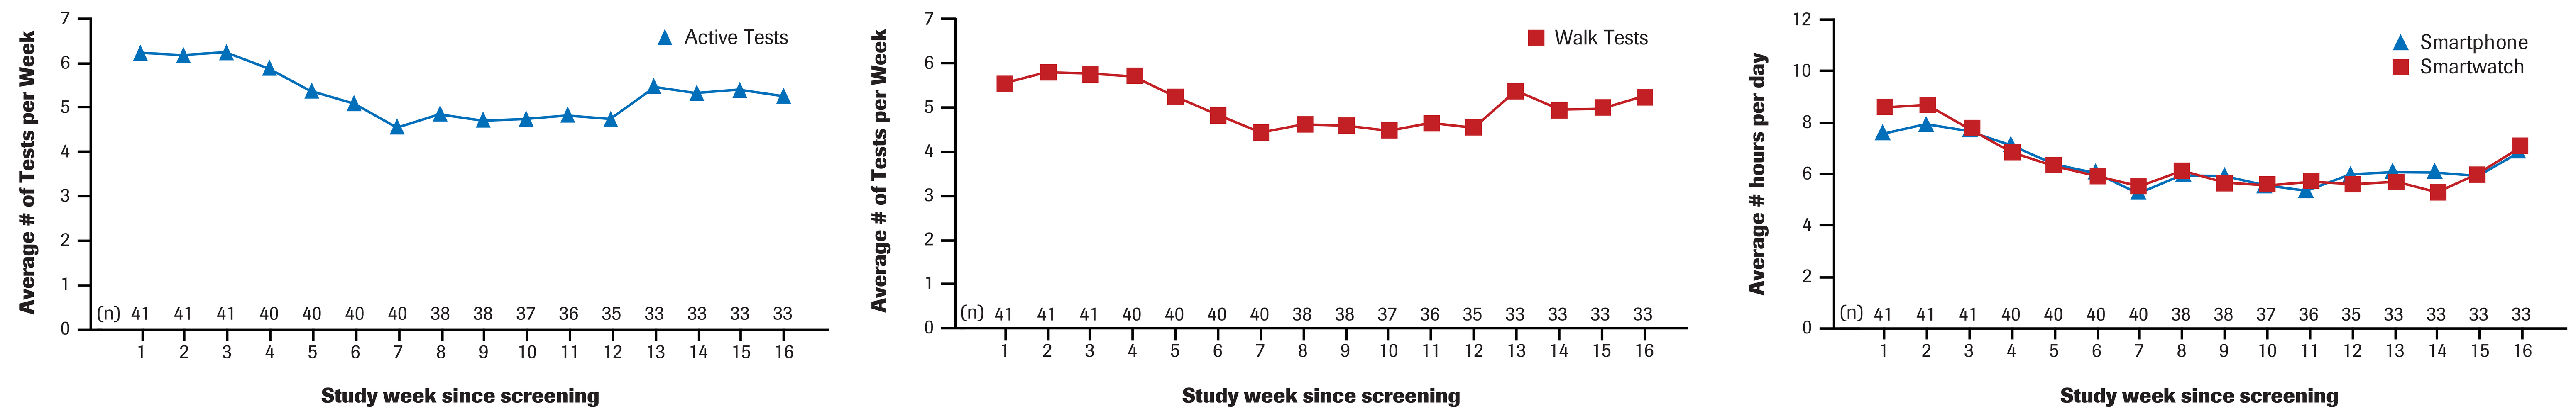
Average # of Tests per Week
Active Tests
Walk Tests
Average # hours per day
Smartphone
Smartwatch
(n) 41 41 41 40 40 40 40 38 38 37 36 35 33 33 33 33
Study week since screening

Average number of active tests, done per study week, excluding the walk test. 3173 active tests have been performed as of data-cut off. Each participant completed an average of 5.28 active tests per week during the first 16 weeks.

Average number of active 2-minute walk tests done per study week. This test can be started independently of the other active tests performed each day. 3022 2-minute walk tests have been performed as of data-cut off. Each participant completed an average of 5.03 walk tests per week during the first 16 weeks.

Average number of smartphone/wearable data recorded per day per study week. Data was considered only if signal thresholds confirmed that phone/watch is carried by the participant. 26778/26882 hours have been recorded as of data cut-off. 6.4 hours of continuous sensor data per day has been collected per patient to date.

Fig 3: The chorea test illustrates the benefits of digital high-frequency monitoring

- This was a proof-of-concept pilot study of a single symptomatic domain of HD.
- The chorea test is designed to detect choreatic movements in the upper limbs.
 - Patients hold the smartphone in their outstretched hand for 30 seconds. They are asked to close their eyes and count down in intervals of 7s from a pre-specified number. The test is completed with the left and the right hand.
- The normalized Acc path (i.e the sum of the euclidean distances between consecutive 3D acceleration signals normalized by test length) was computed using every chorea test performed by a patient from screening to Day 14 of the study.
- Choreatic movements from study participants and independent HCs can be visualised using acceleration data from the chorea test (**Figure 3a**).

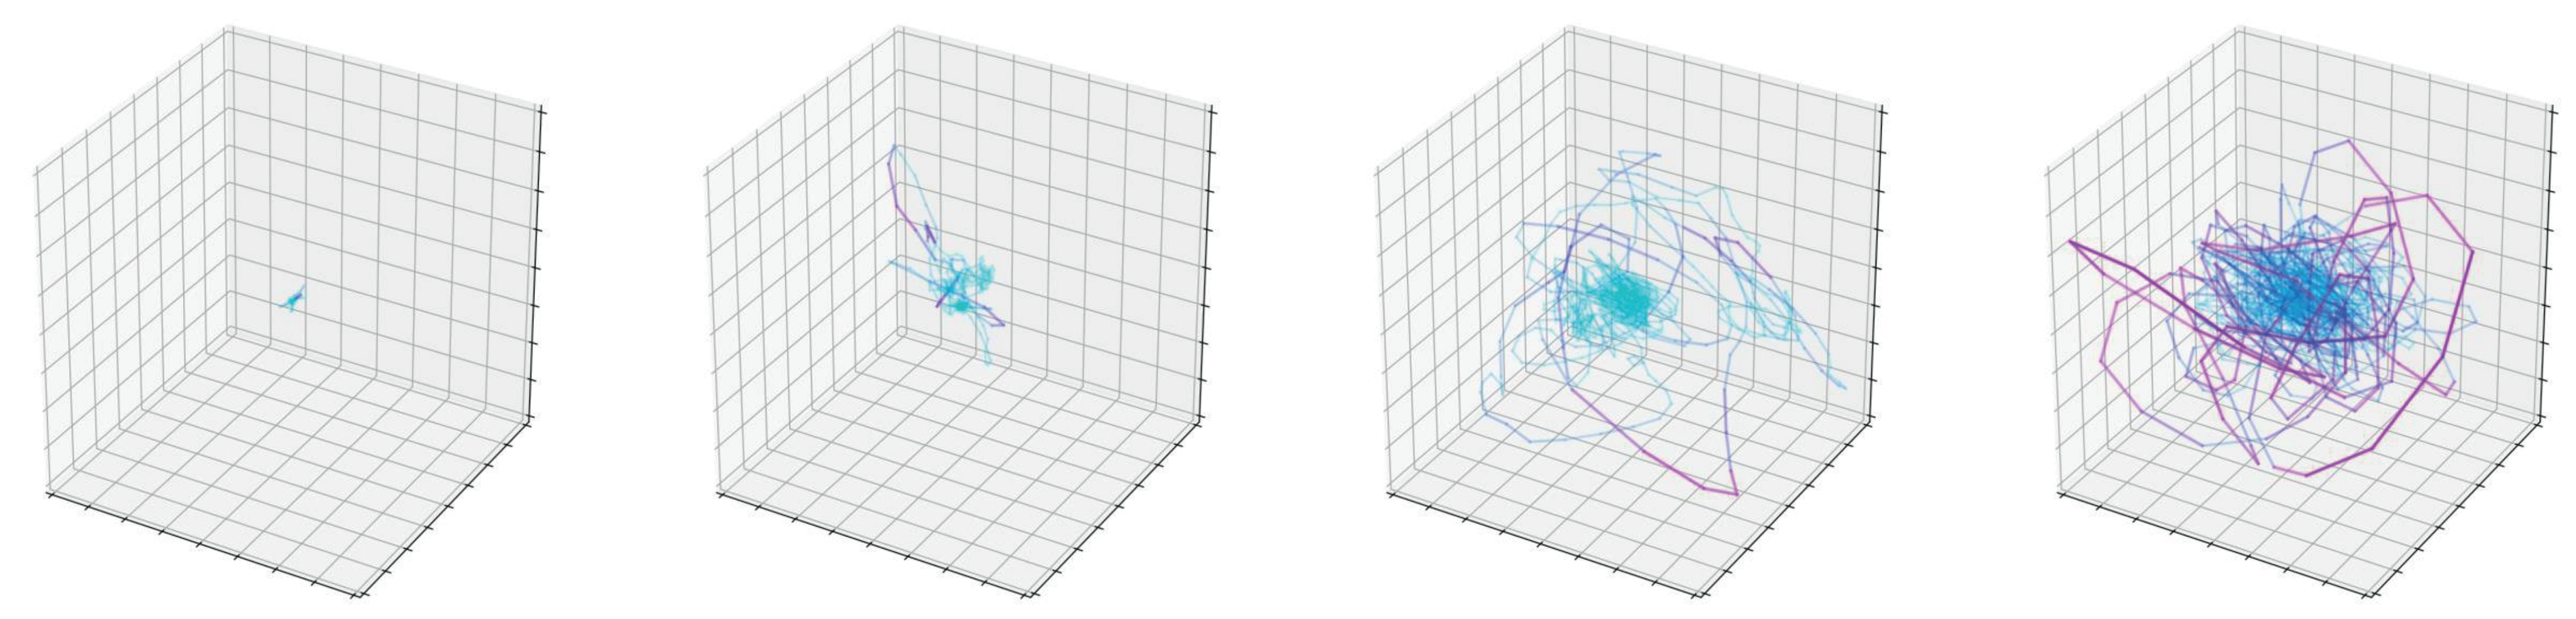

HC | **Subject 1** TMS: 9 | **Subject 2** TMS: 19 | **Subject 3** TMS: 45

Figure 3a. Display of illustrative data collected by patients and a control during the Chorea Test. The charts display 30 seconds of the acceleration path mapped to a 3D plane. The stronger the purple hue is, the faster the movement.

- Analysis of the normalized Acc path from study participants (**Figure 3b**) reveals:
 - correlation with associated chorea items from UHDRS (left hand: r= 0.65 [*P*<0.0001] right hand r=0.49 [*P*=0.001]).
 - the HD group shows increases in normalized Acc Path compared to the independent HC group (left P<0.0001, right P<0.0001).
 - HD subjects with no apparent chorea (chorea upper extremity score=0) on the UHDRS show increases in normalized Acc Path vs the independent HC group (left P<0.0004).

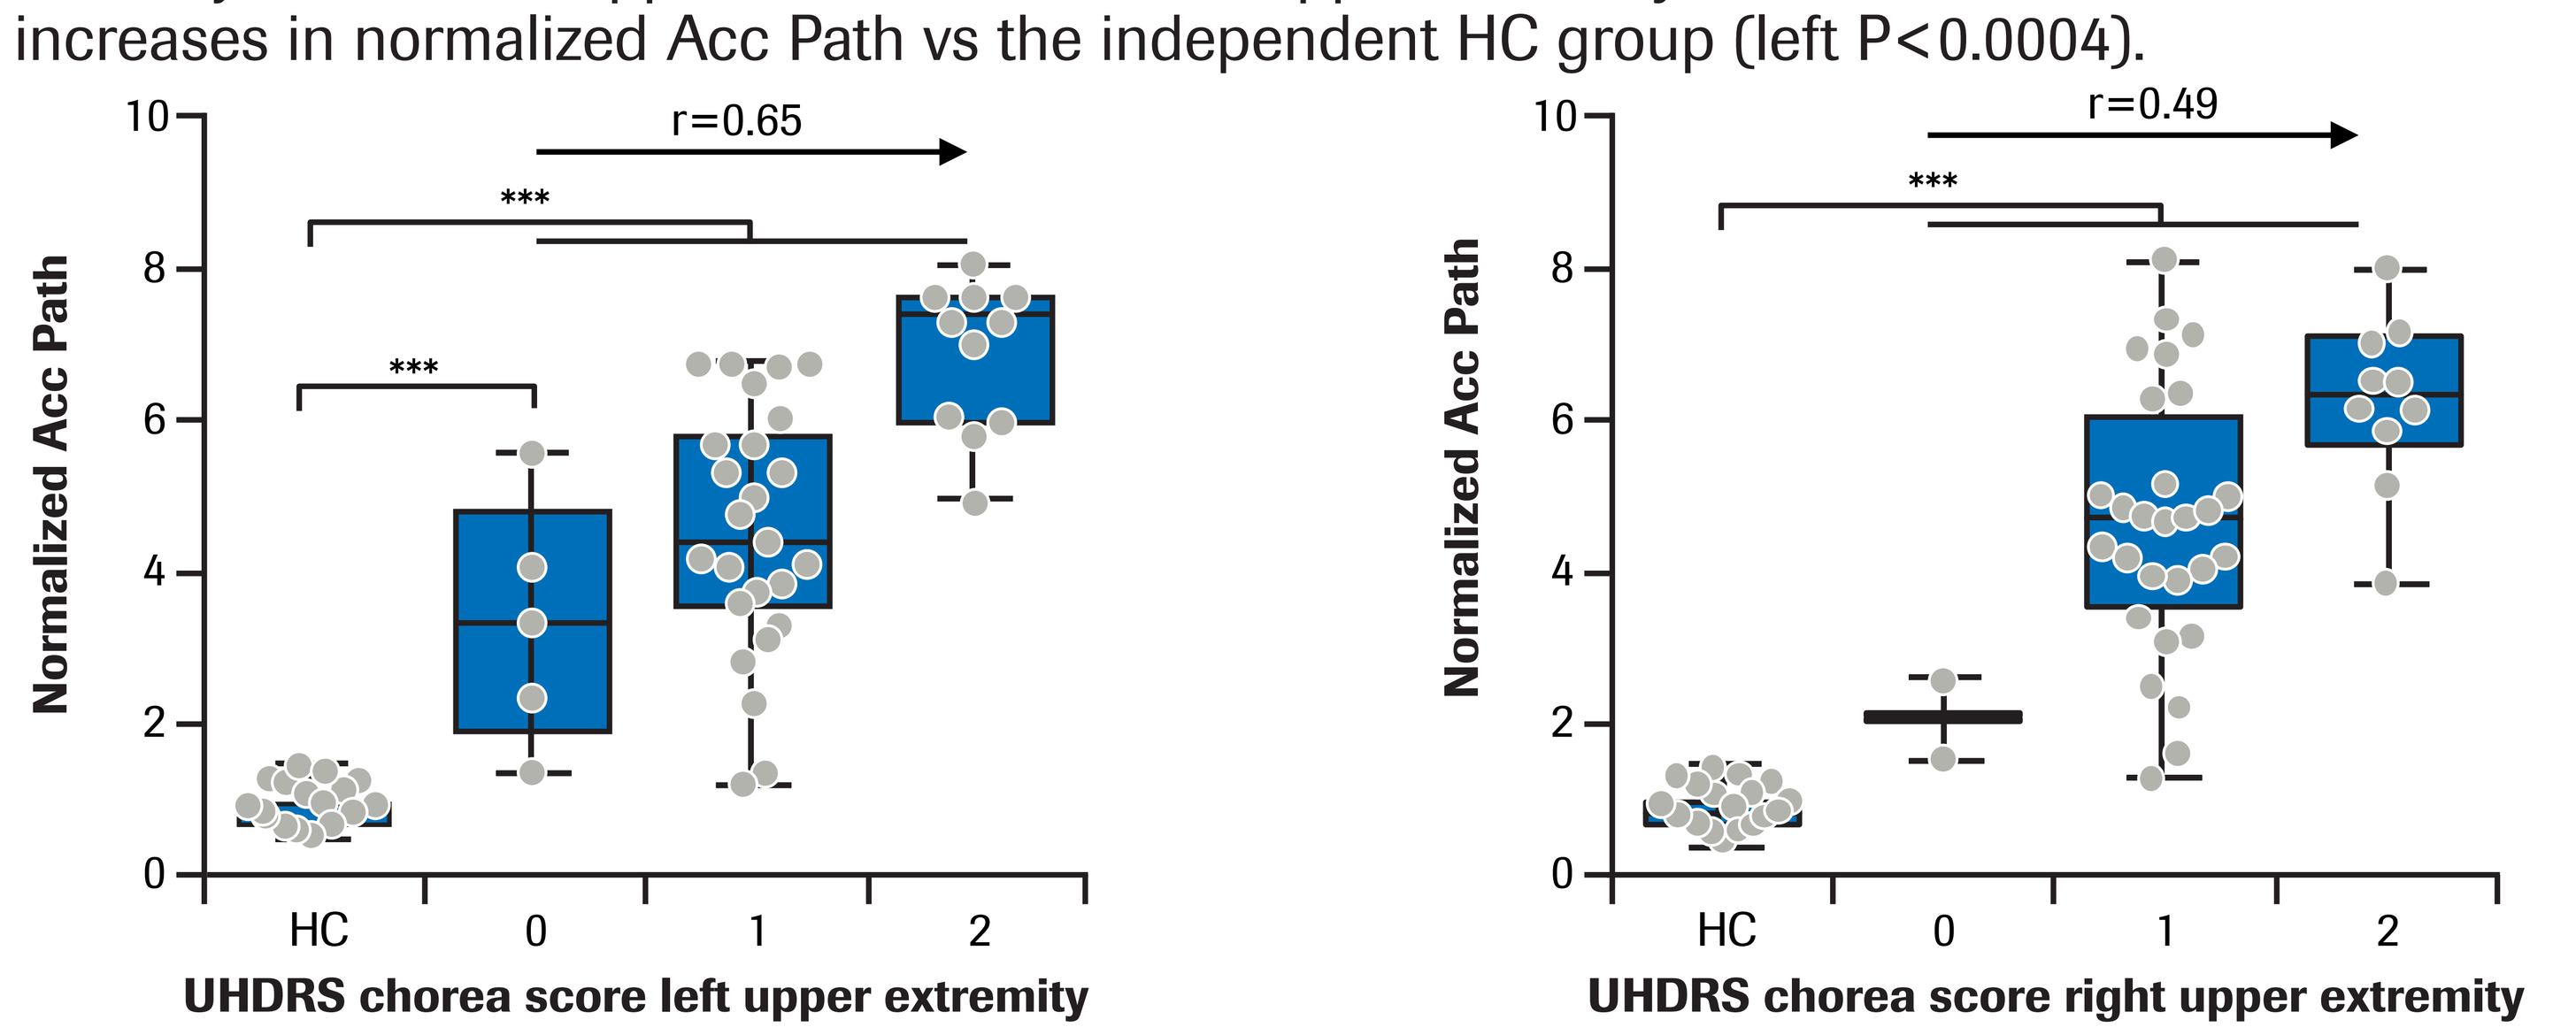
Normalized Acc Path
r=0.65
r=0.49

HC 0 1 2
UHDRS chorea score left upper extremity
UHDRS chorea score right upper extremity

Figure 3b. Analysis of the normalized Acc path from chorea digital monitoring assessments with reference to UHDRS item maximal chorea score.
***= *P*<0.001

Conclusions

- An app-based test suite has been developed to remotely monitor key HD symptoms. Smartphones and smartwatches equipped with this app have been successfully deployed in an ongoing clinical trial.
- Preliminary data suggests that patients adhere to smartphone and smartwatch based assessments, suggesting that these tools may provide an innovative way to learn more about patients' daily symptoms in the clinical trial setting.
 - Preliminary results from the chorea test show good agreement between clinical assessments of chorea severity and read-outs from a smartphone-based chorea test performed at home.
 - Significant differences between an independent control group and patients showing no clinical chorea symptoms suggest a higher sensitivity of the digital test by using motion sensor and daily tests. The relationship of clinical to digital tests under the same clinical testing conditions is being further investigated.
- This was a pilot analysis of a single domain of HD. Further studies will include other domains and also cover measurements from the daily life of a patient such as mobility, chorea events and severity over the day.

Abbreviations

Acc, acceleration; HC, healthy control; HD, Huntington's disease; PRO, patient reported outcomes; SMDT, Symbol Digit Modalities Test; SWR, Stroop Word Reading Test; TFC, Total Functional Capacity; TMS, Total Motor Score; UHDRS, Unified Huntington's Disease Rating Scale.

Acknowledgments

We thank all the patients who participate in our studies and their families. This study was funded by Ionis-Pharmaceuticals and F. Hoffmann La-Roche. The authors thank Craig O'Hare, PhD, of MediTech Media UK for providing editorial support for this poster, which was funded by F. Hoffmann La-Roche in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Huntington's Study Group. *Mov Disord*. 1996; 11:136–142.
2. Lipsmeier F, et al. *Mov Disord*. 2018; 33:1287–1297.

Please scan using your QR reader application to access this poster on your mobile device.
NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details.
Alternatively this can be accessed at: https://bit.ly/2Ptj8lx

Presented at HSG 2018: Unlocking HD, Houston, TX, USA; November 8–10, 2018